Real-World Analysis of C5 Inhibitor Administration Characteristics, Resource Utilization and Total Cost of Care Among Ravulizumab-Treated PNH Patients Using Integrated Medical and Pharmacy Claims

J.P. Burke, PhD[1], D. Eckwright, PharmD[1], K. Broderick, PharmD[2], J. Fishman, PharmD[2], P.P. Gleason, PharmD[1,3]. [1]Prime Therapeutics LLC, Eagan, MN, United States; [2]Apellis Pharmaceuticals, Inc., Waltham, MA, United States; [3]University of Minnesota College of Pharmacy, Minneapolis, MN, United States.

BACKGROUND

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare, potentially life-threatening, chronic condition[1] appearing in any race, gender or age, often diagnosed in the 30s.[3]
- In PNH, blood cells lack complement regulatory proteins, so the body recognizes these healthy red blood cells as damaged leading to uncontrolled activation of the complement cascade causing hemolysis.[1] PNH induced hemolysis can lead to persistently low hemoglobin resulting in frequent transfusions and debilitating symptoms such as severe fatigue and dyspnea.[1,2]
- Hemolysis occurs in PNH through two mechanisms: intravascular hemolysis (IVH), mediated primarily by complement protein C5; and extravascular hemolysis (EVH), mediated primarily by C3.[1]
- At the time of this analysis, treatments included complement C5 inhibitors (C5I) eculizumab (ECU), FDA approved in 2007, and ravulizumab (RAV), FDA approved in 2018, which target IVH by inhibiting C5.[4,5] Bone marrow transplant is the only curative therapy; however, it carries many risks and is not frequently done in PNH.[2] A new therapy, pegcetacoplan, was FDA approved in 2021 and targets both IVH and EVH by inhibiting C3 upstream in the complement cascade.
- PNH treatment goals include reduction in number of transfusions, improvement in hemoglobin levels, and reduction in debilitating symptoms. However, to overcome ongoing hemolysis in patients treated with C5I, clinicians may utilize blood transfusions or increase the dose and/or frequency of C5I beyond FDA label recommendations.[6,7]
- In 2021, eculizumab was identified as the 10th most expensive U.S. drug at $678,392 per year.[8] Since ravulizumab approval in 2018, some clinicians have switched PNH patients from ECU to RAV, which also costs more than $400,000 per year dependent upon patient weight, frequently over $450,000 per year for an adult.[9]
- Given the severity and cost of PNH, it's essential to understand real-world dosing patterns, break through hemolysis, transfusion rates, and PNH total cost of care, especially among those switching from ECU to RAV therapy.

OBJECTIVE

- The primary objectives of this analysis were to use real-world, commercially insured member claims data to:
 - Examine RAV and ECU dosing patterns,
 - Evaluate RAV and ECU effectiveness by characterizing transfusion frequency and break through hemolysis (BTH) episode frequency,
 - Describe real-world total health care costs using administrative claims data,
 - In members switching from ECU to RAV, compare health care costs, BTH and resource utilization before and after switch.

TABLE 1

Maintenance Dosing and Therapy Administration Site of Service During 12-Month Post-Index in Commercial Members* Identified with PNH

	Index Therapy	
Outcome	Eculizumab (n = 34)	Ravulizumab (n = 52)
New to C5I therapy on index date, n (%)	16 (47.2%)	17 (32.7%)
Newly initiating index therapy, n (%)	16 (47.2%)	52 (100.0%)
Duration on therapy, in days, mean (sd)	234.2 (150.4)	301.3 (101.8)
Discontinuation of therapy, n (%)**	18 (52.9%)	18 (34.6%)
Mean time to discontinuation in days, mean (sd)	117.9 (116.1)	180.9 (87.3)
Maintenance therapy – dosing per label-recommended treatment, n (%)	16 (47.0%)	43 (82.7%)
Higher than standard	13 (38.2%)	5 (9.6%)
Lower than standard	0	0
Unknown***	5 (14.7%)	4 (7.7%)
Mean and median time between infusions in days	14.7 (15.0)	60.3 (56.0)
Maintenance therapy – time-between-claims in days, n (%)****		
<=9.99 (ECU)/<=41.99 (RAV)	2 (5.9%)	0
10.00-11.99 (ECU)/42.00-48.99 (RAV)	0	0
12.00-13.99 (ECU)/49.00-55.99 (RAV)	3 (8.8%)	11 (21.2%)
14.00+(ECU)/56.00+(RAV)	24 (70.6%)	37 (71.1%)
Unknown**	5 (14.7%)	4 (7.7%)
Administration of therapy locations, n (%)		
Hospital outpatient	5 (14.7%)	21 (40.4%)
Office	19 (55.9%)	24 (46.2%)
ER	0	0
In-home	2 (5.9%)	4 (7.7%)
Other	8 (23.5%)	3 (5.8%)

* Members were identified between 1/1/2018 through 12/31/2020 and required to have ≥ 6 months of continuous enrollment in the pre-index period, 12 months of continuous enrollment in the post-index period, ≥ 18 years of age at the index date, and no evidence of atypical hemolytic uremic syndrome, generalized myasthenia gravis or neuromyelitis optica spectrum disorder in the pre-index period;
**See Methods for definitions; PNH = paroxysmal nocturnal hemoglobinuria

TABLE 2

Transfusions and Break Through Hemolysis (BTH) During 12-Month Post-Index Period in Commercial Members* Identified with PNH

Characteristics	Eculizumab (n = 34)	Ravulizumab (n = 52)
Transfusions, n (%)		
0	16 (47.1%)	35 (67.3%)
1 – 3	8 (23.5%)	8 (15.4%)
4+	10 (29.4%)	9 (17.3%)
Number of BTH episodes, n (%)		
0	24 (70.6)	45 (86.5)
1	4 (11.8)	2 (3.8)
2	2 (5.9)	2 (3.8)
3+	4 (11.8)	3 (5.8)
Mean number of BTH episodes, mean (sd)	0.9 (2.2)	0.5 (2.0)
Costs per episode, mean (sd)	$18,378 ($42,987)	$8,398 ($18,578)
Total costs of all episodes, mean (sd)	$17,297 ($62,788)	$4,360 ($16,494)
Hospitalization	$16,872 ($62,078)	$4,189 ($16,093)
ER	$425 ($948)	$172 ($676)

* Members were identified between 1/1/2018 through 12/31/2020 and required to have ≥ 6 months of continuous enrollment in the pre-index period, 12 months of continuous enrollment in the post-index period, ≥ 18 years of age at the index date, and no evidence of atypical hemolytic uremic syndrome, generalized myasthenia gravis or neuromyelitis optica spectrum disorder in the pre-index period; PNH = paroxysmal nocturnal hemoglobinuria;
BTH = break through hemolysis. See Methods section for transfusion and BTH identification, and costs calculation specifications.

FIGURE 1

All-Cause Health Care Costs During 12-Month Post-Index Period in Commercial Members* Identified with PNH

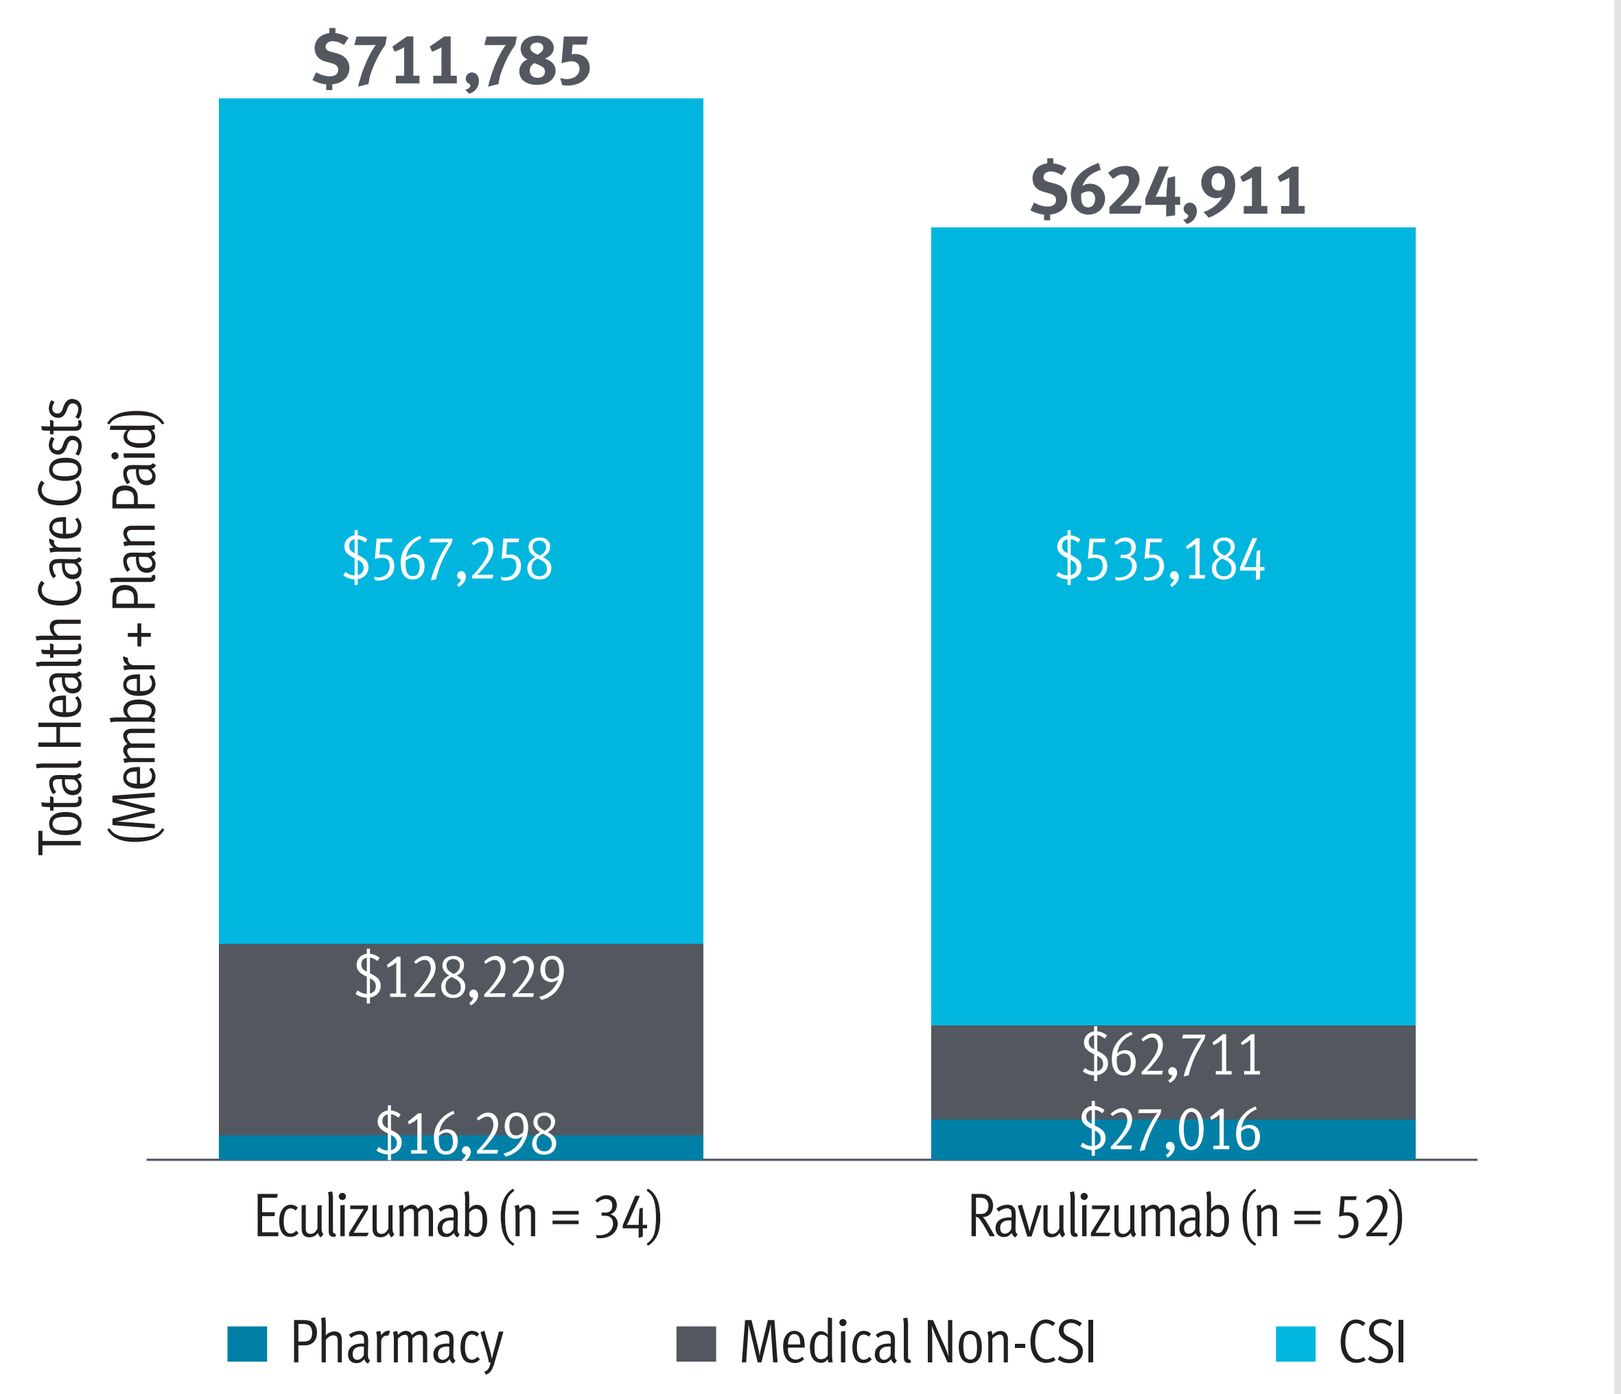
$711,785
$624,911
Total Health Care Costs (Member + Plan Paid)
$567,258
$535,184
$128,229
$62,711
$16,298
$27,016
Eculizumab (n = 34)
Ravulizumab (n = 52)
Pharmacy
Medical Non-CSI
CSI

A sensitivity analysis including only members with a PNH diagnosis was conducted with similar findings: means of $741,472 and $625,338 in the ECU and RAV cohorts, respectively.
* Members were identified between 1/1/2018 through 12/31/2020 and required to have ≥ 6 months of continuous enrollment in the pre-index period, 12 months of continuous enrollment in the post-index period, ≥ 18 years of age at the index date, and no evidence of atypical hemolytic uremic syndrome, generalized myasthenia gravis or neuromyelitis optica spectrum disorder in the pre-index period; PNH = paroxysmal nocturnal hemoglobinuria; C5I = complement 5 inhibitors.

TABLE 3

Health Care Costs, BTH Episodes, and Transfusions in the 6-month Pre-Index and 6-month Post-Index Periods in Commercial Members* PNH who Switched from Eculizumab to Ravulizumab

Outcomes	6-month Pre-Index Period Eculizumab (n = 35)	6-month Post-Index Period Ravulizumab (n = 35)
Pharmacy costs, non C5I, mean (sd)	$1,292 ($5,529)	$1,341 ($6,252)
Medical costs, mean (sd)	$293,012 ($116,352)	$346,516 ($159,209)
C5I costs, mean (sd)	$288,475 ($115,161)	$343,958 ($159,122)
Non-C5I medical costs	$4,537 ($7,557)	$2,557 ($5,029)
Total costs, mean (sd)	$294,304 ($116,831)	$347,857 ($159,122)
Number of BTH episodes, n (%)		
0	33 (94.3)	31 (88.6)
1	0 (0)	1 (2.9)
2	1 (2.9)	1 (2.9)
3+	1 (2.9)	2 (5.7)
Mean number BTH episodes, mean (sd)	0.2 (0.9)	0.4 (1.3)
Costs per episode, mean (sd)	$7,163 ($8,400)	$1,717 ($3,378)
Number of transfusions, n (%)		
0	26 (74.3%)	28 (80.0%)
1–3	5 (14.3%)	4 (11.4%)
4+	4 (11.4%)	3 (8.6%)

*Members who switched from ECU to RAV were identified between 1/1/2018 through 12/31/2020 and required to have ≥ 6 months of continuous enrollment in the pre-index period, 6 months of continuous enrollment in the post-index period with presence of ECU claim in pre-index period and index claim defined as first RAV claim in ID period, ≥ 18 years of age at the index date, and no evidence of atypical hemolytic uremic syndrome, generalized myasthenia gravis or neuromyelitis optica spectrum disorder in the pre-index period; PNH = Paroxysmal nocturnal hemoglobinuria; BTH = break through hemolysis. See Methods section BTH identification and costs calculation specifications.

METHODS

- All analyses were conducted using integrated medical and pharmacy claims data from 15 million commercially insured members.

Identification of Commercial Members with PNH

Members with potential PNH treatment (i.e., claim for ECU or RAV) from Jan. 1, 2018-Dec. 31, 2020 (3-year identification [ID] period) were identified using combined pharmacy and medical claims data. Members were selected based on the following characteristics:

- ≥ 2 ECU infusion claims; the index date was defined as the date of the first ECU infusion
 OR
- ≥ 1 RAV infusion claim; the index date was defined as the date of the first RAV infusion

Note: Members with a claim for both ECU and RAV in the ID period were included in the RAV cohort and their first claim for RAV was considered their index claim.

To be included in the analyses, members must also have had:

- ≥ 6 months continuous enrollment prior to the index date (pre-index period)
- 12 months continuous enrollment after the index date (post-index period)
- ≥ 18 years of age at the index date

Members were excluded based on the following criteria:

- The presence of another indication for ECU or RAV, defined as one or more medical claims with an ICD-10-CM code from the conditions below in any position during the ID period or on index date.
 - Atypical hemolytic uremic syndrome (D59.3)
 - Generalized myasthenia gravis (G70.xxx)
 - Neuromyelitis optica spectrum disorder (G36.0)

To maximize sample size, members with ECU or RAV drug therapy were not required to have a PNH diagnosis.

Outcomes Measurements

- Standard of care treatment dosage and frequency patterns — Compared real-world maintenance dosing to FDA labeled dosing assessed both by dosage (i.e., units of drug administered) and by frequency (i.e., time between claims). A member's standard of care dosage and frequency pattern was calculated through the earliest of discontinuation or end of the study period. Standard of care treatment was not calculated for the three members who switched from RAV to ECU therapy in the post-index period.
 - Based on dosage — Members with a 25% greater or lesser than expected maintenance dose claims were defined as NOT having standard of care treatment. The standard care of treatment was calculated on a mg/day basis while members are on maintenance therapy. For members new to therapy, the maintenance dose was established after accounting for the initial loading doses (5 doses for ECU and 1 dose for RAV). Once a member was on maintenance therapy, they were followed to the earliest of discontinuation date or the end of the study period (365 days). Members were categorized whether they had higher than standard (>80 mg/day), standard (between 80 and 48 mg/day) or lower than standard (<48 mg/day) care treatment. The daily maintenance dose was determined by dividing the total number of units of medication while on maintenance therapy by the number of days on maintenance therapy. This determination was limited for RAV given weight-based dosing as weight information was unavailable. Therefore, the threshold for RAV was set using the highest labeled dose and interval (3,600mg every 56 days). Members were categorized by whether they had higher than standard, standard or lower than standard of care treatment.
 - Based on time-between-claims — Members with time-between-claims lower than expected (i.e., higher frequency dosing) were identified. For members using RAV (56 days expected), time-between claims of <42, 42–48, and 49–55 days were identified. For members using ECU (14 days expected), time-between claims of <9, 10–11, and 12–13 days were identified.
- Transfusions — The number of transfusions in the post-index period was determined. Because of the difficulty in determining the presence of multiple transfusions on a single day using claims data, all transfusions on a single day were considered a single episode. Members were stratified by whether they had 0, 1 to 3, or 4+ transfusions in the post-index period. The time from the index date to the first transfusion and the time between the first and second transfusions were calculated.
- Break through hemolysis (BTH)— Was defined as the presence of one or more ICD-10-CM code(s) for abdominal pain, anemia (excluding aplastic anemia), chronic kidney disease (CKD), dysphagia, dyspnea, erectile dysfunction, fatigue, infection, sepsis, respiratory infection, kidney infection, urinary tract infection (UTI), pulmonary, hypertension, thrombosis in any of the 5 positions on a medical claim and PNH-related hospitalization or ER visit. If BTH was defined via a hospitalization, then the episode was extended through the entire length of the hospitalization. Costs related to BTH were evaluated overall and on a per episode basis. Costs for BTH included medical claims costs related to the ER visits or hospitalizations for claims defined above as BTH.
- Discontinuation — Was defined as an index therapy gap during the post-index period. For ECU (given every 14 days), a member with a gap of more than 28 days between administrations was considered to have discontinued therapy. For RAV (given every 56 days), discontinuation was defined as a member with a gap of more than 112 days between administrations. Members were not considered discontinued if they switched to RAV from ECU within 28 days of their last ECU claim or to ECU from RAV within 112 days of their last RAV claim. Time to discontinuation, duration on therapy, was also determined.
- Health care utilization and site of care — Health care utilization was broken out by hospitalization, emergency room (ER) and office visits. Site of care (office, hospital outpatient, or home infusion) was determined using the medical claim place of service code to categorize place of C5I therapy.
- Health care costs — Health care costs in the 12-month post-index period was determined. The total cost of care included all pharmacy and medical claims inclusive of member share and plan paid allowed amounts. The costs were broken out by medical costs and pharmacy costs. C5I costs were further broken out by site of care.
- Age, gender, proportion of members new to therapy, no claim for a C5I in the pre-index period, and the presence of an ICD-10-CM code for PNH (D59.5) were examined for descriptive purposes.

Statistical Analyses

- Descriptive analyses were used.

RESULTS

Identification of Analytic Population – Commercial Members with PNH

- A total of 452 commercial members with a pharmacy claim for ECU or RAV were identified from Jan. 1, 2018 to Dec. 31, 2020.
- Members meeting PNH criteria among the adult C5I utilizers with 6 months of pre-index and post-index enrollment was 37.2% (97 of 261).
- After applying continuous enrollment and age criteria and excluding members with evidence of another indication, 86 commercial members met analysis criteria: 34 in the ECU cohort and 52 in the RAV cohort.
- The mean age of the members was 42.6 years and 54.6% were female, data not shown.

Maintenance Dosing and Therapy Location During 365 Days Post-Index (Table 1)

- 47.1% (16 of 34) of members in the ECU cohort and 32.7% (17 of 52) in the RAV cohort were naïve to C5I therapy.
- 47.1% (16 of 34) of members in the ECU cohort and 100.0% (52 of 52) in the RAV cohort newly initiated their index therapy, meaning loading doses were much more frequent in the RAV cohort.
- 52.9% (18 of 34) of members in the ECU cohort and 34.6% (18 of 52) in the RAV cohort discontinued therapy. Among those who discontinued, the mean time to discontinuation was 117.9 days in the ECU cohort and 180.9 days in the RAV cohort. Average days of C5I therapy was 234.2 days for ECU and 301.3 for RAV.
- Higher than label recommended dosage, as measured by mg/day, occurred in 38.2% (13 of 34) of ECU members and 9.6% (5 of 52) RAV members. Taking into consideration the highest possible weight-based dose, all exceeded standard dosage.
- When defining maintenance dosing measured by time between claims, 14.7% (5 of 34) of ECU members and 21.2% (11 of 52) of RAV members had mean times between maintenance claims more frequent than labeled dosing.
- The most common site of care for the ECU index claim was the physician's office and for RAV was evenly split between the physician's office and hospital outpatient settings.

Transfusions and Break Through Hemolysis Frequency and Costs During 365 Days Post-Index (Table 2)

- 52.9% (18 of 34) of ECU members and 32.7% (17 of 52) of RAV members had at least one transfusion in the post-index period.
- 29.4% (10 of 34) of ECU members and 17.3% (9 of 52) of RAV members had 4 or more transfusions in the post index period.
- 10 of the 34 ECU members (29.4%) and 7 of the 52 RAV members (13.5%) had at least one BTH episode.
- The mean total costs of all BTH episodes per member was $17,297 in the ECU cohort and $4,360 in the RAV cohort.

All-Cause Health Care Costs and Health Care Utilization During 365 Days Post-Index (Figure 1)

- Mean total health care costs in the post-index period was $711,785 among ECU members and $624,911 among RAV members.
- All C5I claims were billed through the medical benefit and, thus, all costs were included in the medical costs. C5I costs accounted for 79.7% and 85.6% of total health care costs in the ECU and RAV cohorts, respectively.
- C5I costs were $32,074 (5.6%) lower in the RAV cohort.
- The proportion of members with a hospitalization was 38.2% (13 of 34) and 21.2% (11 of 52) in the ECU and RAV cohorts, respectively. (Data not shown)
- The proportion of members with an ER visit was 38.2% (13 of 34) and 23.1% (12 of 52) in the ECU and RAV cohorts, respectively. (Data not shown)

Members in the RAV Cohort with ECU Use in the Pre-Index Period, BTH Episodes, Transfusions and Health Care Costs in the 182 Days Prior to and After Beginning RAV Therapy (Table 3)

- 67.3% (35 of 52) RAV members were identified as switching from ECU to RAV during the pre-index period and had continuous enrollment 182 days prior to and 182 days after their index date (date of switch).
- Mean total health care costs increased from $294,304 in the 6-month pre-index period to $347,857 in the 6-month post-index period. This was driven primarily by C5I costs, which increased from $288,475 in the pre-index period to $343,958 in the post-index period and was likely due to RAV loading dose.
- Two additional members experienced a BTH episode in the post-index period. However, the mean cost per BTH episode decreased from $7,163 (pre-index) to $1,717 (post-index).
- Two fewer members experienced a transfusion in the post-index period.

CONCLUSIONS

Among 15 million commercially insured members meeting analytic criteria, we identified the following real-world C5I treatment findings:

- A concerning 1 in 5 members appears to be receiving a dosage higher than approved by the FDA, 2 in 5 had at least 1 transfusion and 1 in 4 had a BTH episode. Although negative outcomes appear to occur to a lesser extent in the RAV group, these finding potentially indicate inadequate therapy response in a substantial subset of C5I utilizers adding additional therapy costs to an already extremely expensive therapy.
- RAV therapy was associated with a lower discontinuation rate than ECU; however, greater than one-third of members appear to have discontinued therapy during the 12 months post-index potentially indicating the continued need for alternative therapies.
- Ravulizumab utilizers appear to have a mean $87,000 (12 percent) lower total cost of care during the current 12 months post-index compared to ECU. However, this difference should be viewed cautiously due to: 1) new start loading dosage costs, much more frequent in the RAV cohort, and 2) site of care mix, with hospital outpatient drug administration being much for frequent in the RAV cohort. These two differences indicate an even greater potential for total cost of care with RAV therapy.
- Members who switched from ECU to the less frequently dosed RAV experienced a 6-month total cost of care increase of $54,000, likely due to the RAV loading dose.
- Switching to RAV may be associated with higher costs in the short-term due to the loading dose cost; however, this real-world study suggests RAV may be associated with lower discontinuation, health care utilization.
- This real-world, C5I therapy, integrated medical and pharmacy claims data may help inform drug management decisions and optimize drug therapy and outcomes for members receiving these extremely expensive medications.

LIMITATIONS

- Propensity score matching or other balancing was not performed to detect baseline differences among ECU and RAV treated members which may better explain some differences noted between groups.
- Pharmacy claims include assumptions of members' actual drug use and diagnoses.
- Member's dosage was calculated using the dose per day average method calculated over the member's C5I drug post-index period; therefore, member's highest dosage is not independently assessed.
- The data used in this study was limited to a commercial population, and results may not be generalizable to Medicare or Medicaid populations.
- Costs in this analysis are limited to health care claim expenses. However, PNH also results in significant indirect and societal costs.[7]
- The optimal measure of BTH would include laboratory measures of lactate dehydrogenase (LDH) along with clinical symptoms and may not be fully captured in the present analysis given coding limitations.
- 11 of the 34 members in the ECU cohort and 5 of the 52 members in the RAV cohort did not have a medical claim with an ICD-10-CM code for PNH and, thus, their PNH diagnosis is not definite.
- Since individual weights were not available and RAV dosing is weight-based, standard-of-care based on dosage had limited applicability for the RAV cohort.
- ECU requires five loading doses and RAV a single loading dose that increase therapy costs during the induction phase. Therefore, cost comparisons that include members new to therapy should be considered.
- Adherence to therapy was not assessed in the analysis.

REFERENCES

1. Brodsky RA. Paroxysmal nocturnal hemoglobinuria. Blood. 2014;124(18):2804-11. https://doi.org/10.1182/blood-2014-02-522128.
2. Hill A, DeZern AE, Kinoshita T, Brodsky RA. Paroxysmal nocturnal haemoglobinuria. Nat Rev Dis Primers. 2017;3(1). https://doi.org/10.1038/nrdp.2017.28.
3. Canadian Association of PNH Patients. The guide to living well with PNH. 2018. Available from http://www.pnhca.org/wp-content/uploads/PNH_LivingWellGuide_2018_ENG_WEB-FINAL.pdf. Accessed August 10, 2021.
4. Soliris® (eculizumab) [prescribing information]. Boston, MA: Alexion Pharmaceuticals, Inc.; November 2020.
5. Ultomiris® (ravulizumab-cwvz) [prescribing information]. Boston, MA: Alexion Pharmaceuticals, Inc.; June 2021.
6. Cheng WY, Sarda S, Mody-Patel, et al. Real-world eculizumab dosing patterns among patients with paroxysmal nocturnal hemoglobinuria in a US population [paper presentation]. American Society of Hematology Annual Meeting; December 2020.
7. Cheng WY, Sarda SP, Mody-Patel N, et al. Real-world healthcare resource utilization (HRU) and costs of patients with paroxysmal nocturnal hemoglobinuria (PNH) receiving eculizumab in a US population. Adv Ther. 2021;38(8):4461-79. https://doi.org/10.1007/s12325-021-01825-4.
8. McQueen, H. (2021, Sept. 7). The 10 Most Expensive Drugs in the US, Period. GoodRx Health. https://www.goodrx.com/healthcare-access/drug-cost-and-savings/most-expensive-drugs-period.
9. Reuters (2019, Oct. 18). FDA approves Alexion's Ultomiris for another rare blood disease. https://www.reuters.com/article/us-alexion-pharms-fda/fda-approves-alexions-ultomiris-for-another-rare-blood-disease-idUSKBN1WY00B.

4085-A © Prime Therapeutics LLC 03/2022
2900 Ames Crossing Road, Eagan, MN 55121

Academy of Managed Care Pharmacy (AMCP) Annual Meeting, March 30, 2022, Chicago, IL

PATRICK GLEASON, 800.858.0723, ext. 5190
pgleason@primetherapeutics.com

All brand names are the property of their respective owners.

Funding provided by Apellis Pharmaceuticals, Inc.